Evaluation of Two Gingival Crevicular Fluid Sampling Methods

T He*, M Somogyi-Mann, JE Barnes, NB Ramji, XJ Zhou

P&G, Mason, OH, USA

1936

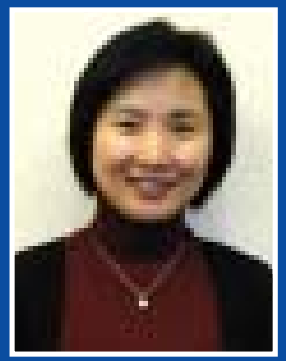

ABSTRACT

Objectives: Quantitative and qualitative assessments of the gingival crevicular fluid (GCF) has been established as a valuable method in assessing cleaning or anti-bacterial performance of oral hygiene products. The purpose of this randomized, 6-period, crossover study was to evaluate the impact of multiple sites versus single site GCF sampling methods in the reduction of within subject variation of total facultative anaerobes (TFAs) concentration in GCF. **Methods**: Twelve healthy subjects were enrolled and completed the study. At baseline and 2 hours post-treatment, 3 GCF samples from distinct sites were collected from either upper left or right quadrant while the opposite upper quadrant was sampled once. Subjects brushed upper right quadrant for 30 seconds, followed by 15 ml of water rinsing for 10 seconds. The same procedure was repeated for upper left quadrant. The samples were placed in dental transport fluid and then plated on enriched tryptic soy agar for TFAs in an anaerobic chamber. Mean change of the TFAs in the GCF from baseline to 2 hours post-treatment were evaluated and Wilcoxon Signed Rank test was used to compare the ratio of the variance of the TFA concentration of the 1 site over 3 sites sampling methods. **Results**:The median of the within subject variation for 3 sites sampling was 0.15 while the variation for the 1 site method was 0.57. The variances of the 1 site and 3 sites sampling methods differed statistically significantly (*p*<0.007). **Conclusions: Collecting GCF samples from 3 sites versus 1 site can reduce the variance of the TFA concentration in GCF.**

INTRODUCTION

Gingival crevicular fluid (GCF) is a key source material for studying the environmental factors associated with periodontal disease. As a serum transudate or inflammatory exudates, GCF contains serum elements, plaque-derived microbial, as well as related host inflammatory response factors. Quantitative and qualitative assessments of the GCF can potentially provide useful information about an individual's oral health status. Previously, this method has been used to assess the interproximal surfactant delivery and cleaning effects of a power toothbrush.

PURPOSE

The objective of the study was to evaluate the impact of multiple sites versus single site GCF sampling methods in the reduction of within subject variation of total facultative anaerobes (TFAs) concentration in GCF.

MATERIALS AND METHODS

Methods:

GCF sampling: performed by placing Periopaper at the entrance of the gingival pocket in mid-interdental area for approximately 30s to absorb the GCF fluid as illustrated.

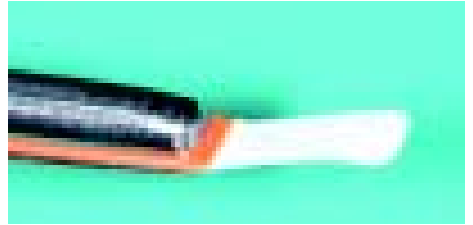

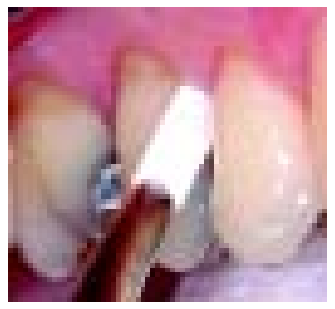

GCF volume was determined using a Periotron®. The sample was then transported in reduced transport fluid, diluted, and plated on enriched tryptic soy agar, and cultured with anaerobic incubation. The number of viable total facultative anaerobes (TFAs) colony forming units (CFU) per ml were subsequently enumerated.

Experimental Design:

This was a six-period cross-over study. Twelve healthy subjects were enrolled into the study. At baseline, 3 GCF samples from distinct sites were collected from either upper left or right quadrant while the opposite upper quadrant was sampled once. GCF samples from the 3 sites were pooled and plated on one plate. Subjects brushed the upper right quadrant with 0.25g of toothpaste (0.23% NaF) for 30 seconds, rinsed with 15 ml of water for 10 seconds. The procedure was repeated for the upper left quadrant. GCF sampling was collected 2 hrs post treatment at the correspondent site(s).

Statistics:

Variance of the change from baseline TFA concentration in GCF was calculated for the 1 and 3 sites methods for each subject and variance ratio was derived**.** Wilcoxon Sign test was used to assess whether the ratio is greater than one**.**

Repeated measures model for cross-over design was also used to model the change from baseline TFA concentration and to estimate the corresponding variances. The model included the baseline concentration as a continuous covariate, sampled sites and period as class variables, and subjects as a random class variable. The model used heterogeneous compound symmetry to estimate the 1 and 3 sites variances.

RESULTS

- The variation of the 1 site sampling method was nearly 3 times larger when compared to the 3 sites sampling method (*p*<0.02). Table 1.

TABLE 1. DESCRIPTIVE SUMMARY
VARIANCE OF CHANGE FROM BASELINE TFA CONCENTRATION - LOG$_{10}$ (CFU/ì L)

Sampling Methods	N[a]	Median	Mean (SD)	Min. - Max.
1	12	0.57	0.60 (0.354)	0.15-1.34
3	12	0.15	0.26 (0.337)	0.07-1.28
Variance Ratio[b]	12	2.87	5.12 (5.086)	0.44-15.96

[a]Number of subjects. [b]Variance ratio of the 1 site variance over the 3 sites variance.

- The modeled variance estimate was 0.34 for 1 site and 0.16 for 3 sites (Table 2). This model fits statistically significant better than the model which assumes equal variances (*p*<0.01).
- The mean reduction of TFA in GCF was relatively similar between the two methods (*p*=0.62). Table 2

TABLE 2. RELATED MEASURES MODEL
CHANGE FROM BASELINE TFA CONCENTRATION

Sampling Methods	N[a]	Adjusted Mean	Variance	Methods Comparison *p*-value
1	12	-0.49	0.340	0.621
3	12	-0.44	0.159	

[a]Number of subjects.

CONCLUSION

- Collecting GCF samples from multiple sites versus single site can reduce the variance of the TFA concentration in GCF.
- The viable bacterial measure from Periopaper® collected from interproximal GCF represents a simple and effective way to assess interproximal cleaning.

MARCH 9-12, 2005

P&G

Research presented at the 83rd General Session of the IADR, March 9-12, 2005