Salivary Peroxide Kinetics with 6% Hydrogen Peroxide Whitening Strips

D.A. McMillan[1], C. Mahony[2]*, L.B. Patel[1], X. Zhou[1], R.E. Bailey[1], G.L. Walden[1]

1046

[1]Procter & Gamble, Mason, OH, USA, [2]Procter & Gamble, Egham, UK

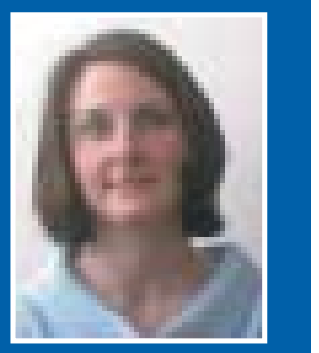

ABSTRACT

Objective: Previous research has demonstrated low salivary exposure to peroxide following use of peroxide-containing vital bleaching systems. This clinical trial was conducted to evaluate the salivary kinetics of strip-based, 6% hydrogen peroxide gel used for vital bleaching. **Methods**: A total of 12 subjects (mean age = 36 years) applied 6% hydrogen peroxide strips to the maxillary arch for 5, 10, 30 and 60 minutes. Salivary samples were collected, and then, strips were removed. All samples (including pretreatment) were analyzed immediately after collection using validated auto-titration methods. Peroxide concentrations below the level of detection were recorded as “0”. **Results**: At baseline, the median peroxide concentration was 0. After strip use, median peroxide concentrations in saliva were 0.017%, 0.004%, and 0.006%, and 0.002%, at 5, 10, 30, and 60 minutes respectively. These low salivary peroxide levels during and after treatment are consistent with those previously reported for other strip and tray-based systems. Overall, median salivary HP concentrations were less than 0.02% at any time point measured, and the maximum level detected did not exceed 0.1% peroxide. Mean Area Under Concentration-Curve (AUC) through 30-minutes of exposure was 0.227 ± 0.089 units (% * minutes). **Conclusion**: <u>This clinical research confirms the rapid degradation of hydrogen peroxide and minimal oral and systemic exposure following use of 6.0% hydrogen peroxide whitening strips</u>.

INTRODUCTION

Many tooth bleaching products exist in what is a growing whitening market, either via dentists or direct to consumers. An important measure of safety is the oral exposure to peroxide during the course of tooth whitening. This poster describes the results from a clinical study evaluating the hydrogen peroxide concentration in the saliva during use of Crest® Whitestrips®.

MATERIALS AND METHODS

Product tested:

Crest Whitestrips[i]; A polyethylene film coated on one side with 6% HP gel (12 mg HP).

[i]*The Procter & Gamble Company, Cincinnati, OH, USA.*

Study design

This was a supervised, cross-over 12 subject trial. Only maxillary teeth were treated and each subject completed a single dosing-time point/day with four time points. The sampling times were 5, 10, 30 and 60 minutes (recommended CWS use is for 30 minutes). A baseline saliva sample was also taken. Prior to dosing, subjects brushed with Crest Cavity Protection.

Sample Analyses

All samples were analyzed immediately after sample collection. At the appropriate sample time, a saliva sample was collected in a cryovial. The device was then removed. Acid was added immediately to the sample and analysed using a PeroXOquant Quantitative Peroxide Assay (Pierce), which is an indirect colorimetric analysis (limit of detection = 0.27 µg/mL).

RESULTS

	HP% in saliva 0 mins	HP% in saliva 5 mins	HP% in saliva 10 mins	HP% in saliva 30 mins	HP% in saliva 60 mins
Median	BDL	0.017	0.004	0.006	0.002
Min-Max	BDL-0.0002	0.006-0.04	0.002-0.01	0.002-0.02	BDL-0.018

DATA

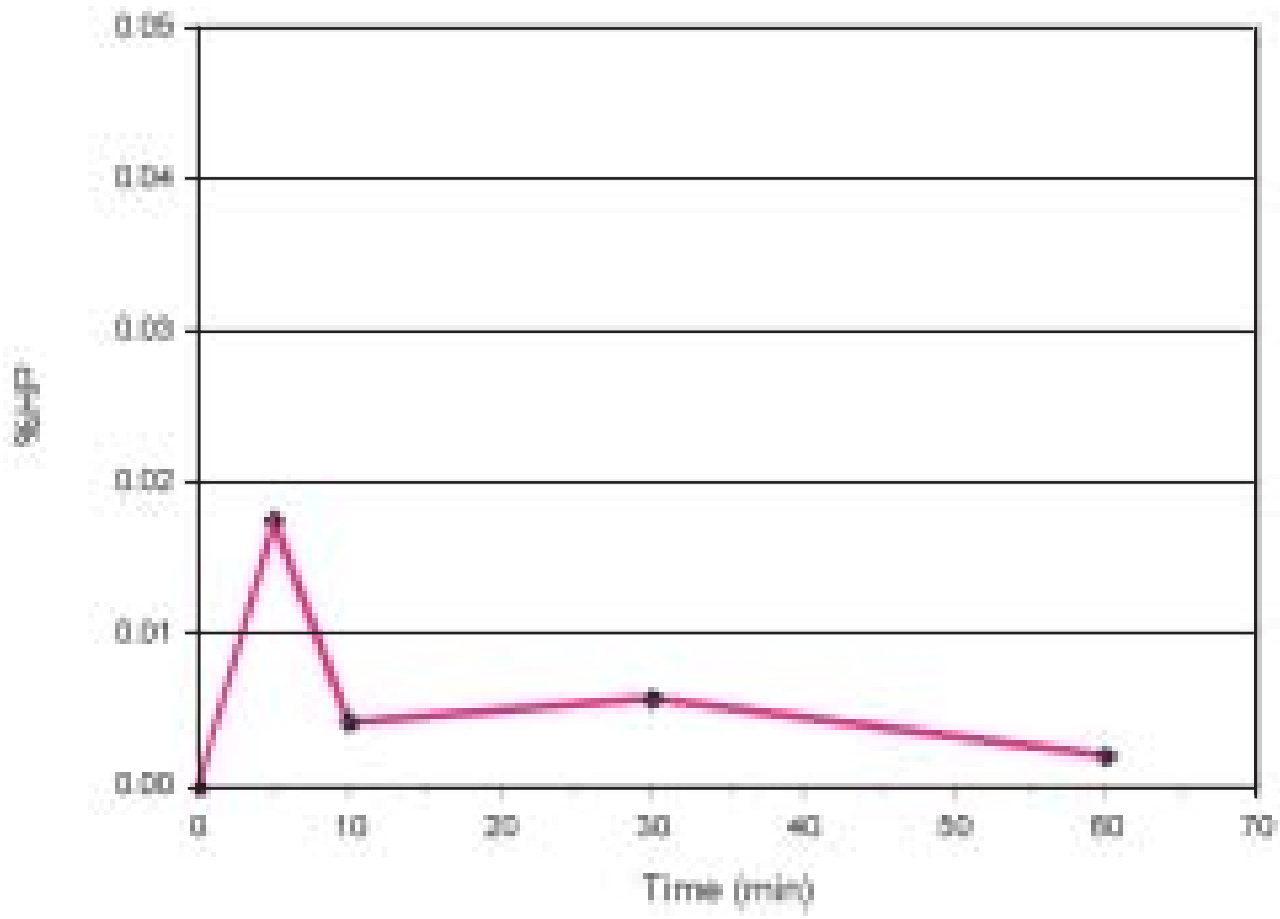
0.05
0.04
0.03
0.02
0.01
0.00
%HP
0
10
20
30
40
50
60
70
Time (min)

CONCLUSION

This clinical research confirms the safety of the current marketed Crest Whitestrips (6.0% hydrogen peroxide strip, 12 mg HP) by demonstrating the low concentration of HP during strip use. This results in very low salivary HP levels and minimal oral exposure (as measured by AUC).

Research presented at the 81ST General Session of the IADR, June 25-28, 2003

Procter&Gamble