A New *in vitro* Caries Prevention Model — Dose Response Study

J.V. Hoskins, M.A. Kaminski*, S.L. Eversole, R.V. Faller
P&G, Mason, OH, USA

3539

ABSTRACT

Fluoride containing dentifrices function through mechanisms that provide both reversal of current lesions as well as inhibition against lesion initiation. Most *in vitro* model systems incorporate both mechanisms of action, but are unable to differentiate between the relative contribution of each factor individually. In addition, most *in vitro* methods are time consuming and labor intensive. **Objectives:** The purpose of this study was to determine applicability of a rapid, inexpensive *in vitro* model focused solely on the protective nature of fluoride. **Methods:** The dose response to fluoride using NaF/silica based formulations {a. 2800 ppm F (NaF)/silica; b. 1100ppm F (NaF)/silica; c. 250ppm F (NaF)/silica; and d. placebo (<1ppm F)} was determined using standard surface microhardness techniques (Vicker's diamond). Each test cell consisted of 3 cores removed from human tooth crowns that were subsequently ground and polished to remove the fluoride rich natural surface. The resultant virgin enamel was soaked in pooled human saliva for 24 hours (freshened 3 times) to develop an early pellicle. Each group of specimens was subjected to a 30-minute treatment in a 1:1 slurry (w:w) of the appropriate toothpaste, rinsed with deionized, distilled water, then subjected to a demineralization solution (0.1M/L Lactic acid, 0.2% Carbopol, 50% saturated w/respect to HAP, pH 5.0) for 2 successive periods of 2 hours each. After each demineralization period, surface measurements (VHN) were taken (200g load). **Results:** Mean change in hardness values for each group [2hr(S.D.)/4hr(S.D.)] were: a) [-24.7(7.7)/-33.3(7.4)]; b) [-23.1(6.1)/-54.4(14.0)]; c) [-53.7(5.8)/-82.3(12.1)]; and d) [-67.1(14.7)/-98.8(23.5) with $a \geq b \geq c \geq d$ ($p<0.05$) at the 4 hour time point. **Conclusions: These data suggest a rapid *in vitro* screening model is capable of demonstrating differences in the protective nature of fluoride in line with the clinical performance of these same products. This model can be completed in approximately 5% of the time required for more traditional in vitro assessment methods and should be considered when assessing fluoride based formulations.**

INTRODUCTION

The use of pH cycling models to demonstrate potential anticaries activity of fluoride containing formulations provides a relatively straightforward way for manufacturers to confirm the efficacy of formulations – either those being marketed or those under development. As currently practiced, these models routinely incorporate features that include both remineralization as well as demineralization aspects of the caries process. Thus, although net efficacy can be predicted with current models, they do not allow discrimination between the portion of the net benefit that is provided through remineralization processes vs. those provided through an inhibition of the demineralization process.

This model focuses solely on the level of protection afforded the enamel after treatment, followed by acidic challenges that represent demineralization processes.

PURPOSE

The purpose of this study was to determine applicability of a rapid, inexpensive *in vitro* model focused solely on the protective nature of fluoride.

MATERIALS AND METHODS

Test products included in this study included: **2800 ppm F** (NaF); **1100ppm F** (NaF); **250ppm F** (NaF)/silica; **Placebo** (<1ppm F). All formulations were prepared with the same, fluoride compatible silica abrasive.

Each test cell consisted of 3 cores removed from human tooth crowns that were ground and polished to remove the fluoride rich natural surface. The resultant virgin enamel was soaked in pooled human saliva for 24 hours (freshened 3 times) to develop an early pellicle. Each group of specimens was exposed, under constant agitation, to a 30-minute treatment in a 1:1 slurry (w:w) of the appropriate toothpaste, then rinsed with deionized, distilled water. Following treatment, each group of specimens was subjected to 25ml of demineralization solution (0.1M/L Lactic acid, 0.2% Carbopol, 50% saturated w/respect to HAP, pH 5.0) for 2 successive periods of 2 hours each. After each demineralization period, surface hardness measurements were taken using a Leitz Miniload Tester and a Vicker's diamond at at constant load of 200g.

RESULTS AND DISCUSSION

While both the remineralizing benefits and the ability of fluoride to inhibit against demineralization are well known, most of the previous work has focused on the ability of low levels of fluoride to inhibit demineralization processes when exposed simultaneously to an acidic environment. This work focused on the ability of fluoride containing dentifrices to deliver a "protective layer" to the tooth surface to provide enhanced tooth resistance against acidic challenges. Based on the results of this study, the use of conventional fluoride containing dentifrices (1100ppm F) provides an enhanced level of protection against demineralization that lasts for several hours.

Higher concentration dentifrice (2800ppm F) provided an enhanced level of protection that lasts longer than that provided by conventional level products, and a reduction in fluoride below current levels suggests a decreased level of long term protection against demineralization delivered from each brushing. These results are consistent with human caries clinical studies that demonstrate enhanced anticaries efficacy for products containing higher levels of fluoride, and a potential for reduced anticaries efficacy for dentifrices containing significantly lower levels of fluoride.

The model system, as described, can be completed in roughly 5% of the time required for conventional pH cycling models. Coupled with data from other model systems, this mechanistic specific model helps provide fast and inexpensive insights into a specific formulation's ability to provide protection against demineralization processes, separate from its overall ability to provide a net caries benefit.

Research presented at the 80th General Session of the IADR, March 6-9, 2002

P&G

3539

A New *in vitro* Caries Prevention Model — Dose Response Study

J.V. Hoskins, M.A. Kaminski*, S.L. Eversole, R.V. Faller
P&G, Mason, OH, USA

Pg. 2

Comparison of Rapid Caries Prevention Model to the Standard pH Cycling Model

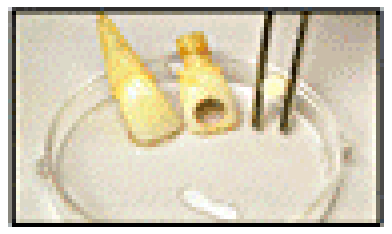

Ground and polished to remove fluoride rich outer surface

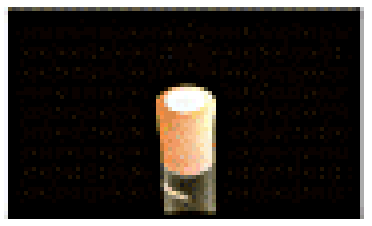

Standard pH Cycling Model

96 hr Carbopol lesion

Treatment Schedule

1 hr. saliva bath (initial pellicle formed)
1 min treatment (1:3 slurry of dentifrice:saliva)
1 hr. saliva bath
1 min treatment (1:3 slurry of dentifrice:saliva)
1 hr. saliva bath
3 hr. exposure to demineralizing solution
1 hr. saliva bath
1 min treatment (1:3 slurry of dentifrice:saliva)
1 hr. saliva bath
1 min treatment (1:3 slurry of dentifrice:saliva)
saliva bath (overnight)

Repeat cycle for 6 days

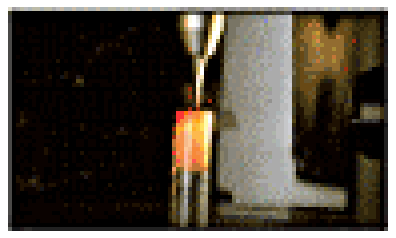

72 hr Demin solution

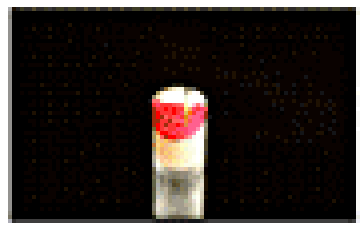

Transverse Micro Radiography

Treatment = 10 days
Analyses = 10 days

Rapid Caries Prevention Model

3 hr Pooled saliva (refreshed 3x)

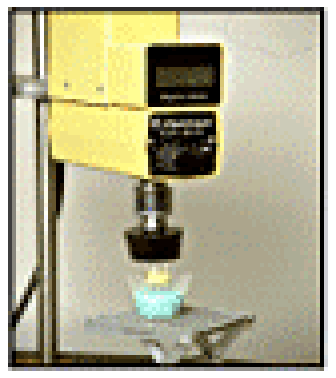

30 min Treatment

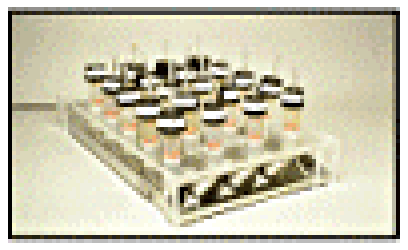

2 hr Demineralization

Repeat

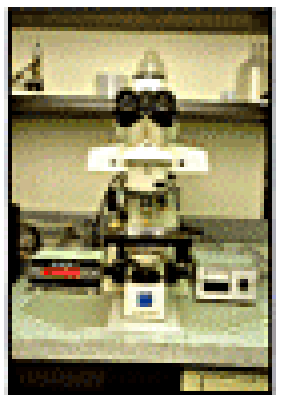

Surface hardness

Treatment and Analyses = 1 day

DATA

Mean Change in Enamel Hardness

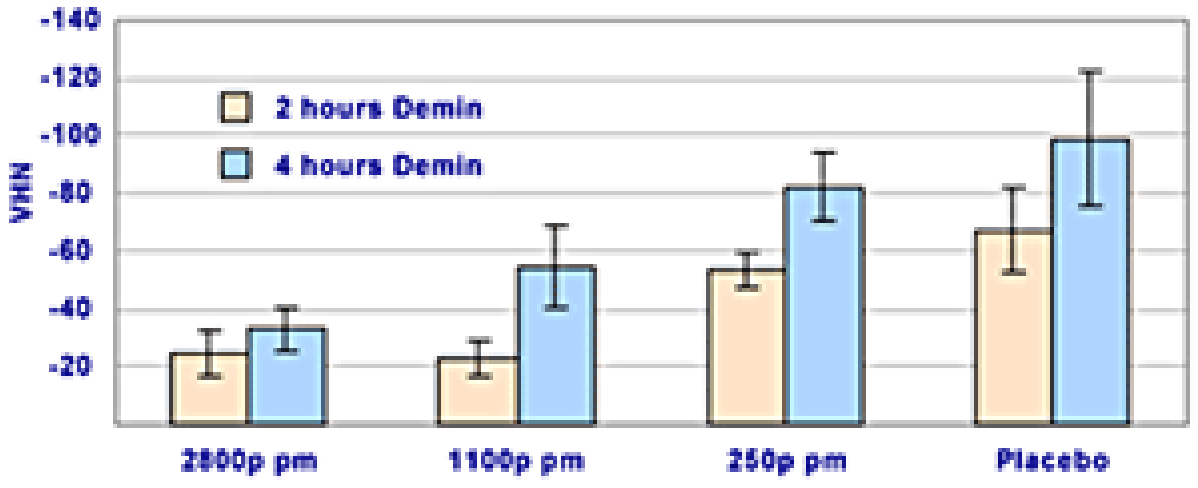
-140
-120
-100
-80
-60
-40
-20
VHN
2 hours Demin
4 hours Demin
2800p pm
1100p pm
250p pm
Placebo

Dentifrice Treatment

CONCLUSION

These data suggest a rapid *in vitro* screening model is capable of demonstrating differences in the protective nature of fluoride in line with the clinical performance of these same products.

Research presented at the 80th General Session of the IADR, March 6-9, 2002

P&G