Comparative Efficacy of Two Mouthrinses in a 6-Month Study

J. Witt[1], R. Gibb[1], A. Hamilton[1], N. P. Lang[2], A. Albert-Kiszely[2],
B. E. Pjetursson[2], G. E. Salvi[2], and G. R. Persson[2]
[1]P&G, Mason, OH, USA, [2]University of Berne, Switzerland

2053

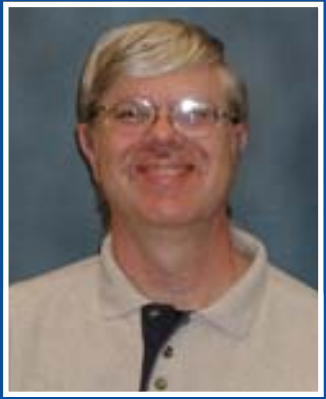

ABSTRACT

Objective: The objective of this study was to compare the effects of a commercial CPC (cetylpyridinium chloride) mouthrinse containing 0.07% CPC (Crest ProHealth Rinse) versus those provided by a commercial essential flavor oil mouthrinse (listerine Antiseptic) on dental plaque accumulation and prevention of gingivitis in an unsupervised 6 month clinical study. **Methods:** This was a double blind, 6-month, parallel group, positive controlled study involving 128 subjects who were balanced and randomly assigned to either positive control (essential oil) or experimental (CPC) mouthrinse treatment groups. The CPC mouthrinse passed proposed performance assays by the FDA for an OTC CPC mouthrinse. At baseline, subjects received a dental prophylaxis and began unsupervised rinsing twice daily with 20 ml. of their assigned mouthwash for 30 seconds after brushing their teeth for 1 min. Subjects were assessed for gingivitis and gingival bleeding by the Gingival Index (GI) of Loe-Silness and plaque by the Silness and Loe Plaque Index (PI) at baseline and after 3 and 6 months of product use. Oral soft tissue health was also assessed. Microbiological samples were also taken for community profiling by the DNA-DNA checkerboard method. **Results:** Results show that after 3 and 6 months use there was no significant difference (p = 0.05) between the CPC and essential mouthrinse treatment groups for overall gingivitis status, ginval bleeding, and plaque. At 6 months the covariant (baseline)-adjusted mean GI and bleeding site numbers for the CPC and essential oil mouth rinses were 0.52 and 0.53 and 5.5 and 6.3, respectively. Both mouth rinses were well tolerated by the subjects. Microbiological community profiles were similar for the 2 treatment groups. **Conclusion**: This study shows that the 0.07% CPC mouthrinse can provide similar plaque and gingivitis benefits to those provided by an essential oil mouthrinse over a 6 month period.

PURPOSE

The objective of this study was to compare the anti-gingivitis and anti-plaque efficacy of a properly formulated, no-alcohol, 0.07% CPC mouthrinse to a mouthrinse containing essential oils as its active system over a 6 month period of time.

MATERIALS AND METHODS

This was a randomized, double-blind, longitudinal (6 month) parallel group, positive-controlled study. Planned measurements at baseline, and after 3 and 6 months of product use included assessments for gingivitis and gingival bleeding sites by the Gingival Index (GI) of Loe-Silness, plaque by the Silness-Loe Plaque Index (PI), and Oral Soft Tissue Health (OST) by a visual examination of the Oral Cavity. Eligible subjects had to have a minimum of 18 gradeable, uncrowned natural teeth with 4 molars, a whole mouth GI score between 0.4 and 1.0. A total of 128 subjects finished the study. After grading for all parameters at baseline, subjects were provided a dental prophylaxis and were randomly assigned and balanced to one of 2 mouth rinse treatment groups: 0.07% CPC or essential oil (positive control). Subjects were instructed to rinse unsupervised with 20 ml of their assigned mouthrinse treatment for 30 seconds twice daily for the next 6 months. Prior to use of the mouthrinse subjects were instructed to brush their teeth for 1 min with a supplied NaF dentifrice (Vivid White) followed by expectoration and a subsequent water rinse. Prior to study start, the 0.07% CPC mouthrinse was tested and passed proposed performance tests prescribed by the FDA for an OTC CPC mouthrinse.
The demographics and balance for the various treatments for subjects completing the study are shown below.

Baseline Study Demographics and Balance for Subjects Completing 6 Months

Mouth Rinse	N	Male/ Female	Smoker/Non Smoker	Mean Age	Mean GI	Mean Bleeding Sites	Mean Plaque
0.07% CPC	65	18/47	15/50	39.8	0.81	15.3	0.44
Essential Oil	63	17/46	16/47	39.1	0.76	13.0	0.41

3 and 6 Month Covariance Adjusted Study Results

Treatment	Gingivitis (GI)			# Bleeding Sites			Plaque (PI)		
	Mean	2-Sided Confidence Interval	2-Sided p-value	Mean	2-Sided Confidence Interval	2-Sided p-value	Mean	2-Sided Confidence Interval	2-Sided p-value
3 Month Results									
0.07% CPC	0.51	(-0.04, 0.03)	0.851	6.9	(-2.16, 0.43)	0.270	0.31	(-0.01, 0.07)	0.211
Essential Oil	0.51			7.7			0.38		
6 Month Results									
0.07% CPC	0.52	(-0.06, 0.04)	0.656	5.5	(-2.07, 0.49)	0.306	0.27	(-0.05, 0.03)	0.785
Essential Oil	0.53			6.3			0.28		

RESULTS

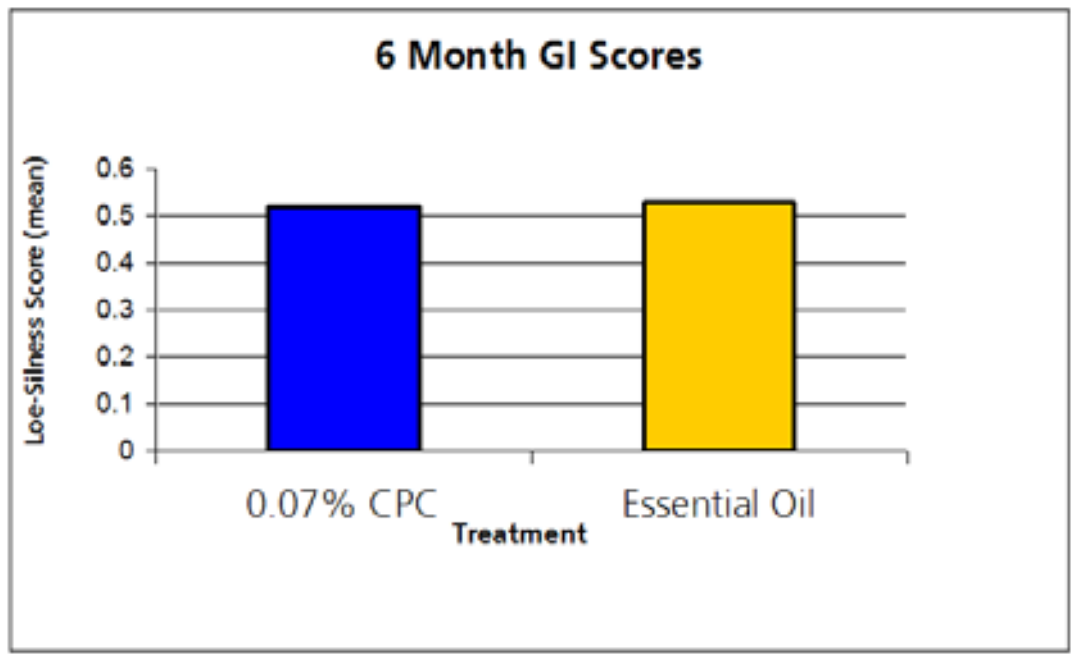
6 Month GI Scores
Loe-Silness Score (mean)
0.6
0.5
0.4
0.3
0.2
0.1
0
0.07% CPC
Essential Oil
Treatment

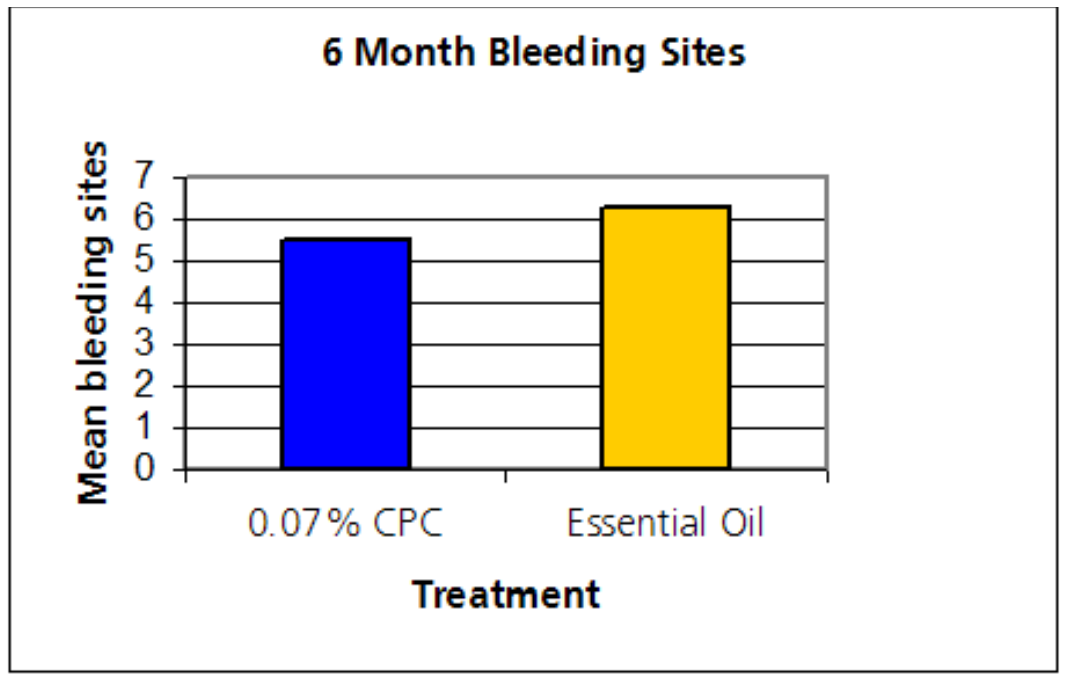
6 Month Bleeding Sites
Mean bleeding sites
7
6
5
4
3
2
1
0
0.07% CPC
Essential Oil
Treatment

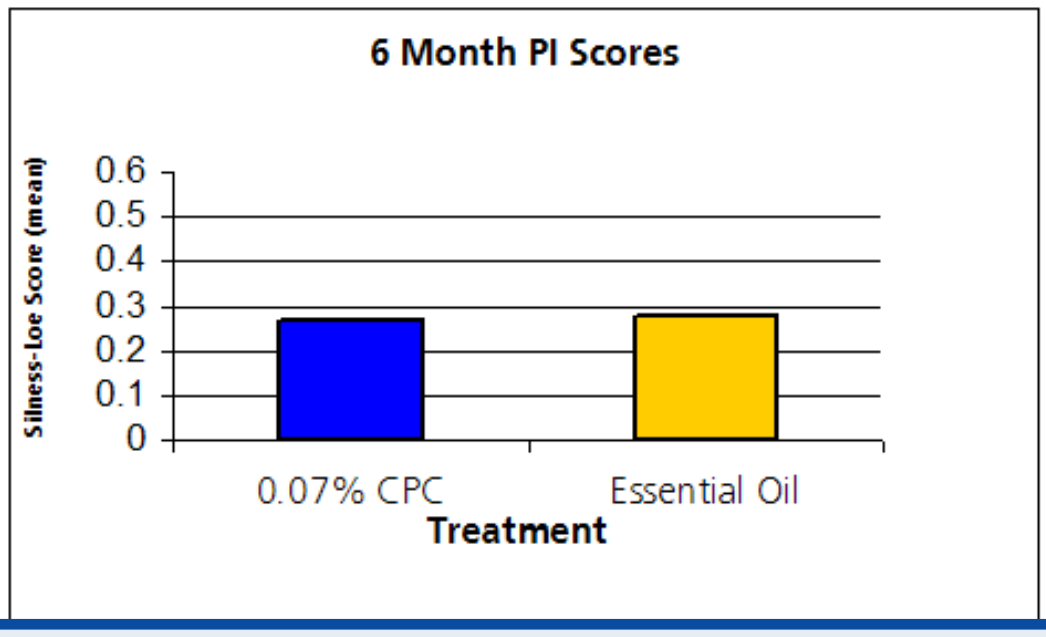
6 Month PI Scores
Silness-Loe Score (mean)
0.6
0.5
0.4
0.3
0.2
0.1
0
0.07% CPC
Essential Oil
Treatment

CONCLUSION

- This study shows that a no-alcohol, 0.07% CPC mouthrinse can provide plaque and gingivitis benefits similar to those of an essential oil mouthrinse over a 6 month period.

MARCH 8-11, 2006

P&G

Research presented at the 84th General Session of the AADR, March 8-11, 2006